(CR-013) CLINICAL EFFICACY OF A FULL-FUNCTION LIGHTWEIGHT AND PORTABLE NEGATIVE PRESSURE WOUND THERAPY (NPWT) WOUND CARE SYSTEM APPLIED ON VARIOUS WOUND TYPES

Claus Brandigi, M.D.[1], Neha Amin, D.O. [1], Zaheed Hassan, M.D. [1], Bounthavy Homsombath, M.D. [1], Beretta Craft-Coffman, PA-C [1], Shawn P. Fagan, M.D. [1], Tayseer Chowdhry, M.D. [1], Joan Wilson, MSN, MHA, RN [2], Amanda Mifflin, BSN, RN,CWOCN [3], Katie Lovesy, RN, BSN [3], Austin Price, CRC II [2], Kimberly Middleton, CRC II [2], Pat Schwab [4]

[1] Burn and Reconstructive Centers of America | [2] Joseph M. Still Research Foundation | [3] Wellstar Cobb Hospital | [4] Medela, LLC

INTRODUCTION

Prospective non-randomized dual-center interventional study evaluates the use of a full-function personal pump* with dual lumen suction drain** in an advanced wound care clinic setting.

The “goal of therapy” for each individual patient was determined at the beginning of treatment - i.e. decrease wound volume in order to progress for closure by split thickness skin graft etc. Therefore the use of the NPWT pump was to bridge the patient to the next level of care and not necessarily to close the wound within the relatively short 4 week study duration.

METHODS

- Patients treated in the outpatient clinic and the burn operating room, or at bedside, then discharged soon after.
- Primary objective: Determine acceptable progress towards the goal of therapy when treating multiple wound types when using a NPWT system* during a 4-week study duration as defined by the physician upon initial assessment.
- Twenty-five patients were included with a mean age of 51.2 (SD: 18.2), 14 were men (56%), 14 were African American (56%), 10 were Caucasian (40%), and 2 were Pacific Islander (4%).
- Wound indications ranged from: Diabetic Foot Ulcers, Pressure Injuries/Pressure Ulcers, surgical, acute/traumatic and dehisced wounds, together with various anatomical locations such as lower extremities, abdominal, flank, and back injuries.

	Baseline (n=25)		W4 (n=25)		Change (n=25)	
	Mean (SD)	Median [IQR]	Mean [SD]	Median [IQR]	Mean (SD)	Median [IQR]
Wound Surface	51.6 (40.0)	54.0 [18.9, 66.0]	25.9 (21.6)	19.3 [8.3, 41.9]	-25.7 (38.1)	-13.2 [-34.0, -1.0]
Wound Volume	174.2 (221.0)	94.1 [13.7, 165.0]	36.3 (63.6)	8.4 [3.9, 34.8]	-138.0 (216.2)	-41.6 [-123.0, -10.5]

Table 1: Wound surface and volume at baseline, at week 4 and the change in mean or median between these time points. IQR: interquartile range.

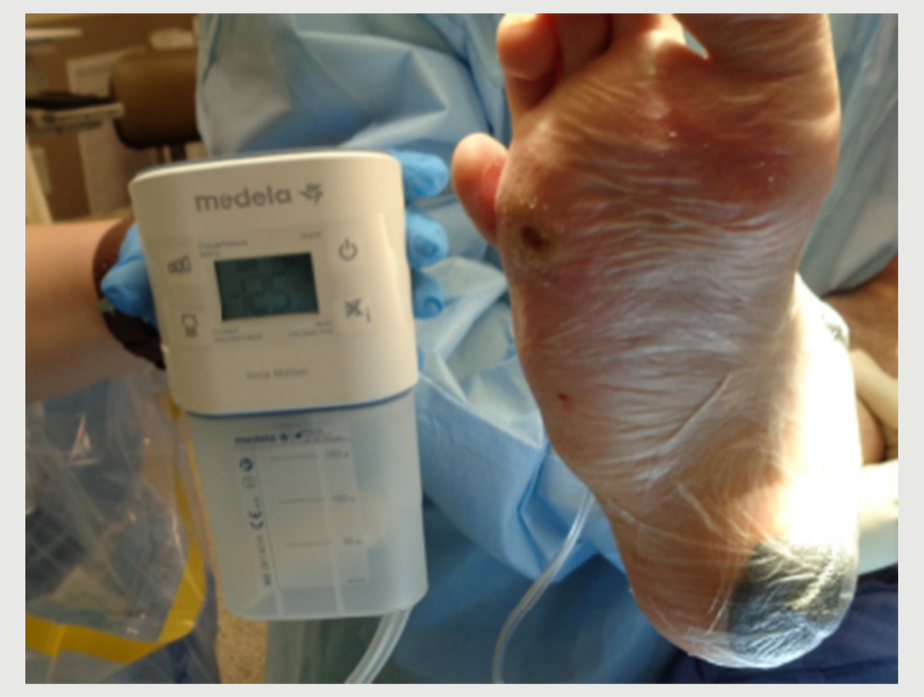
medela

Patient with ultra portable NPWT system* with dual lumen suction drain**

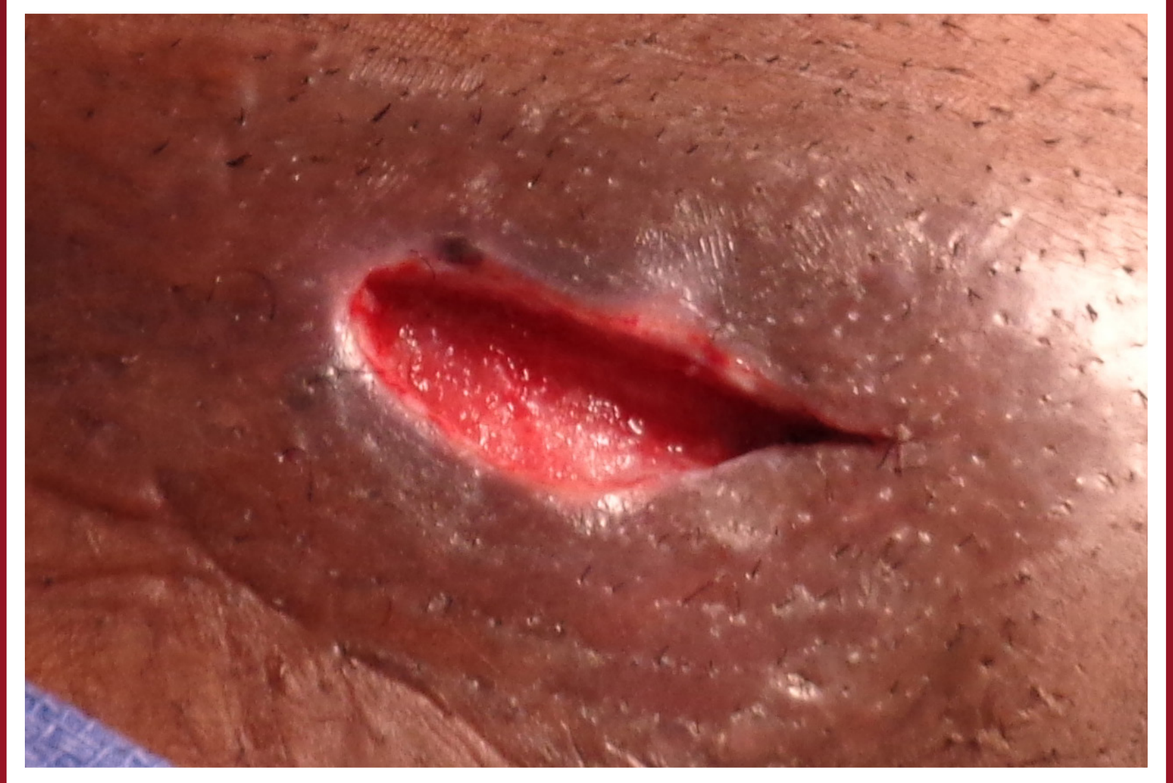

Groin wound week 1

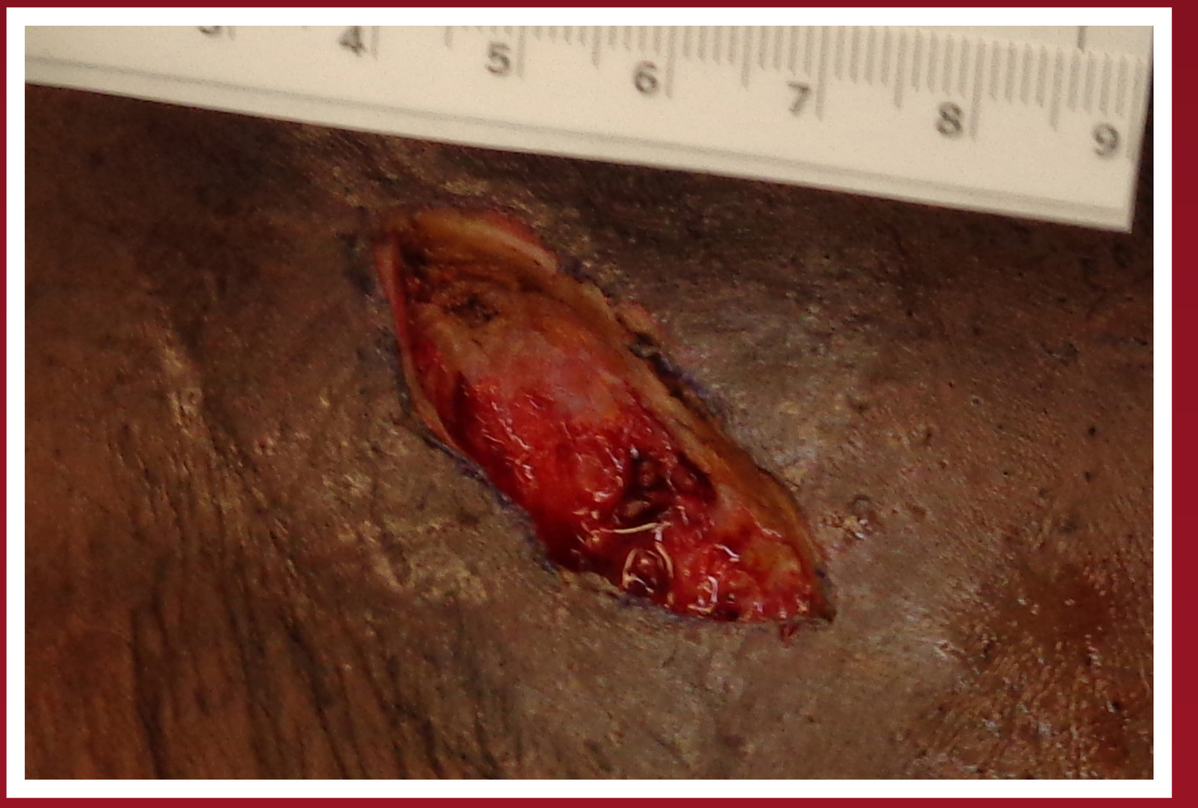

Groin wound week 2

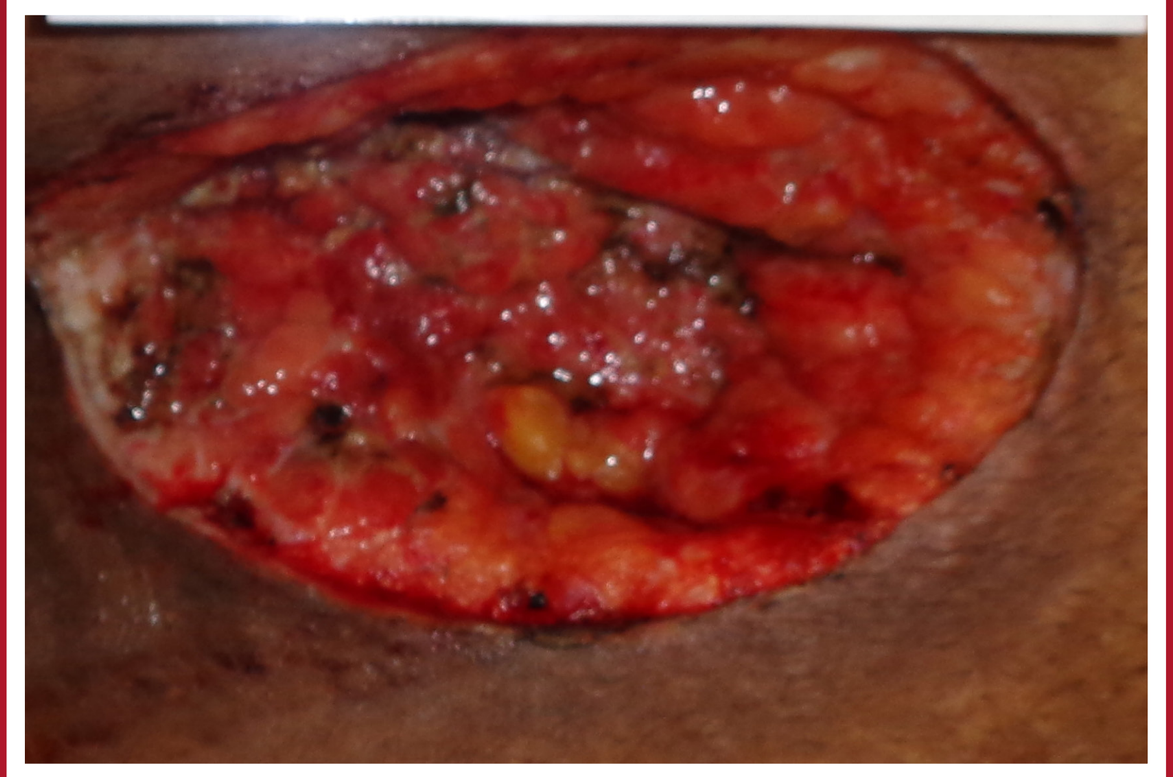

Flank wound baseline

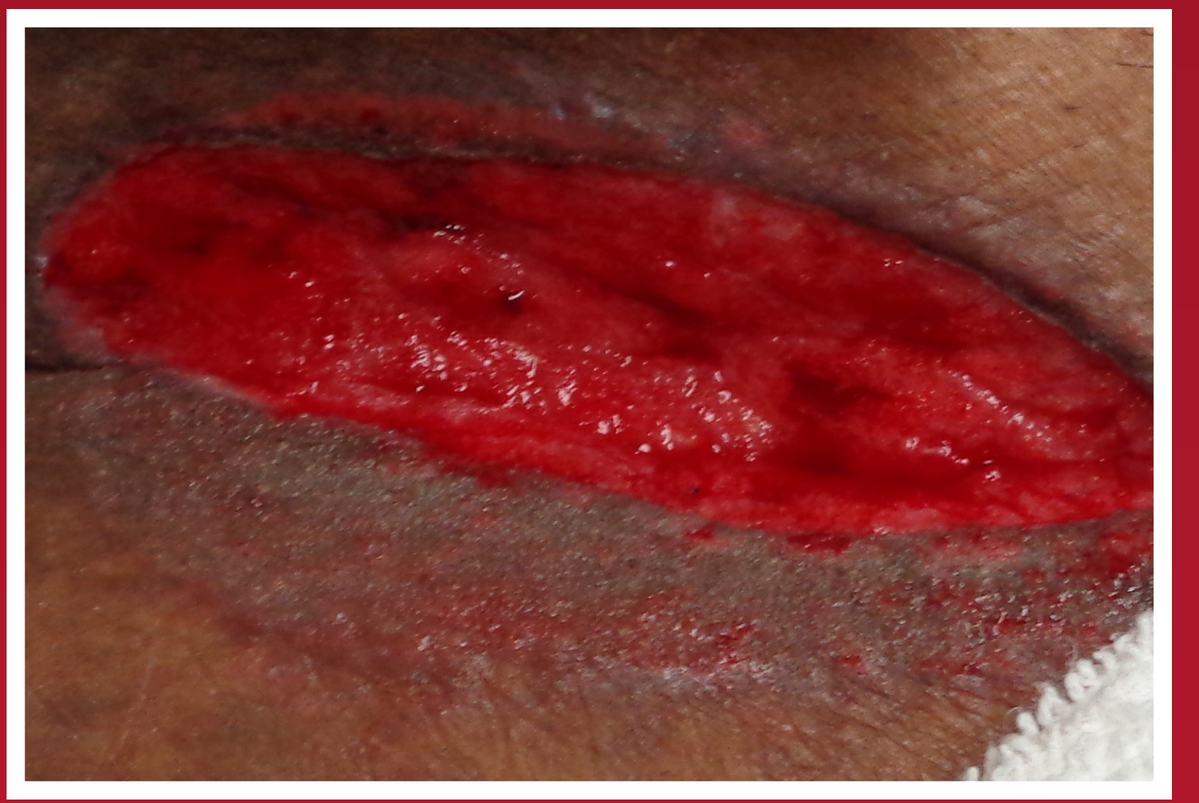

Flank wound week 3

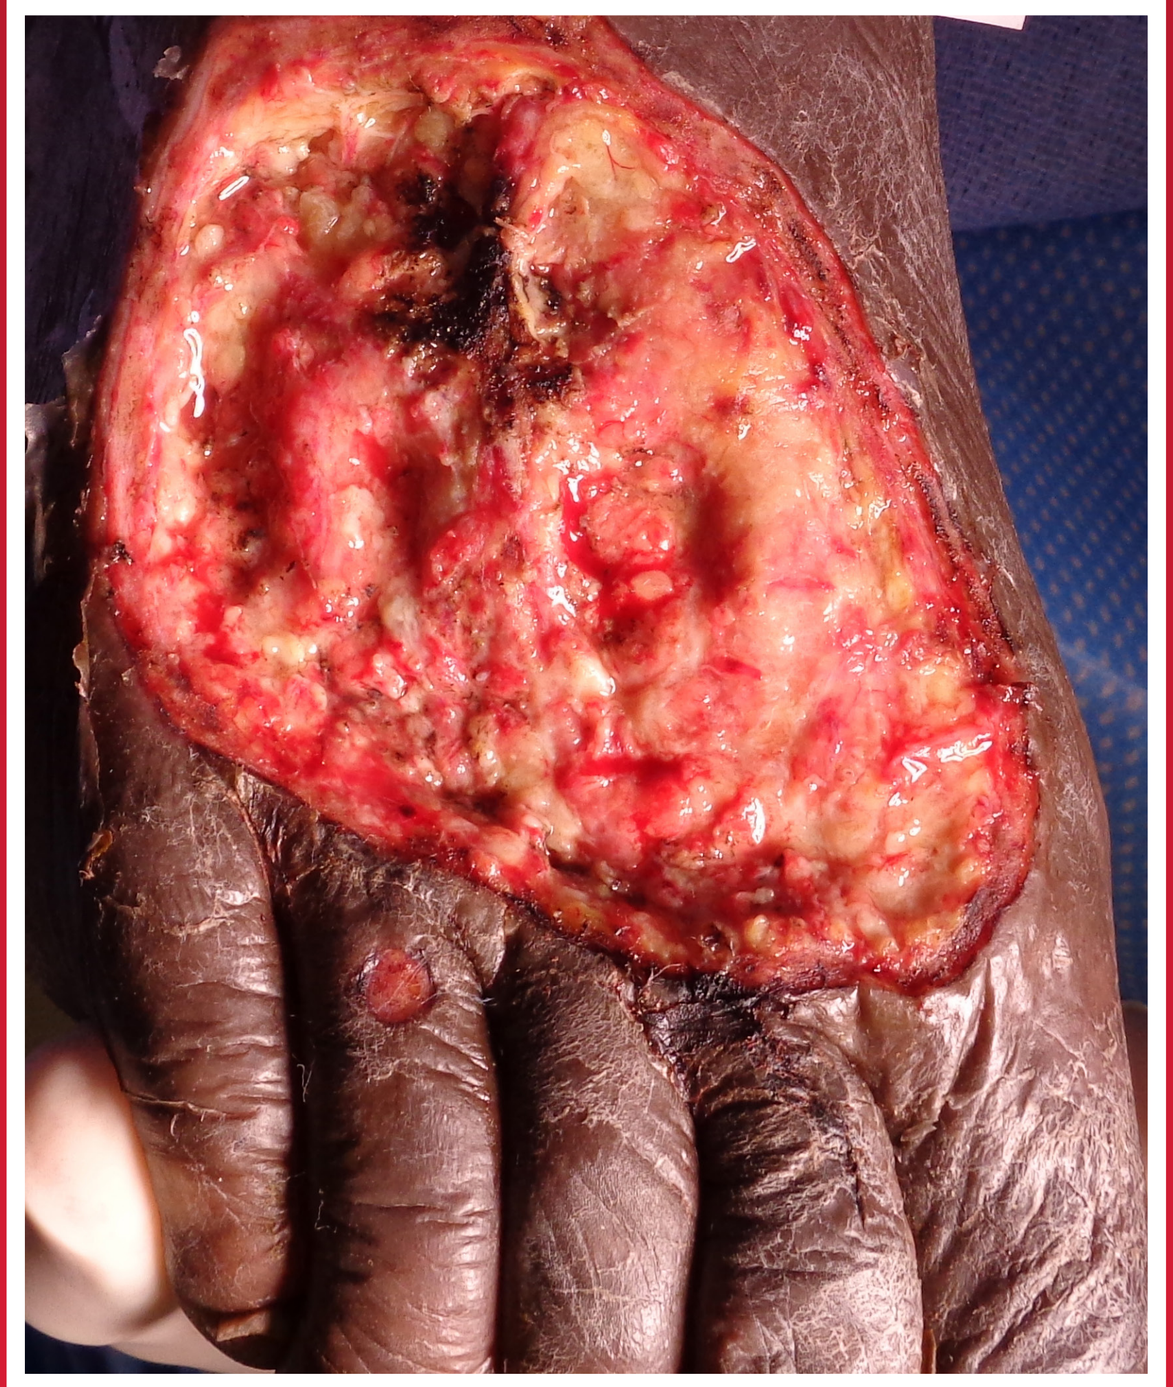

Foot wound baseline

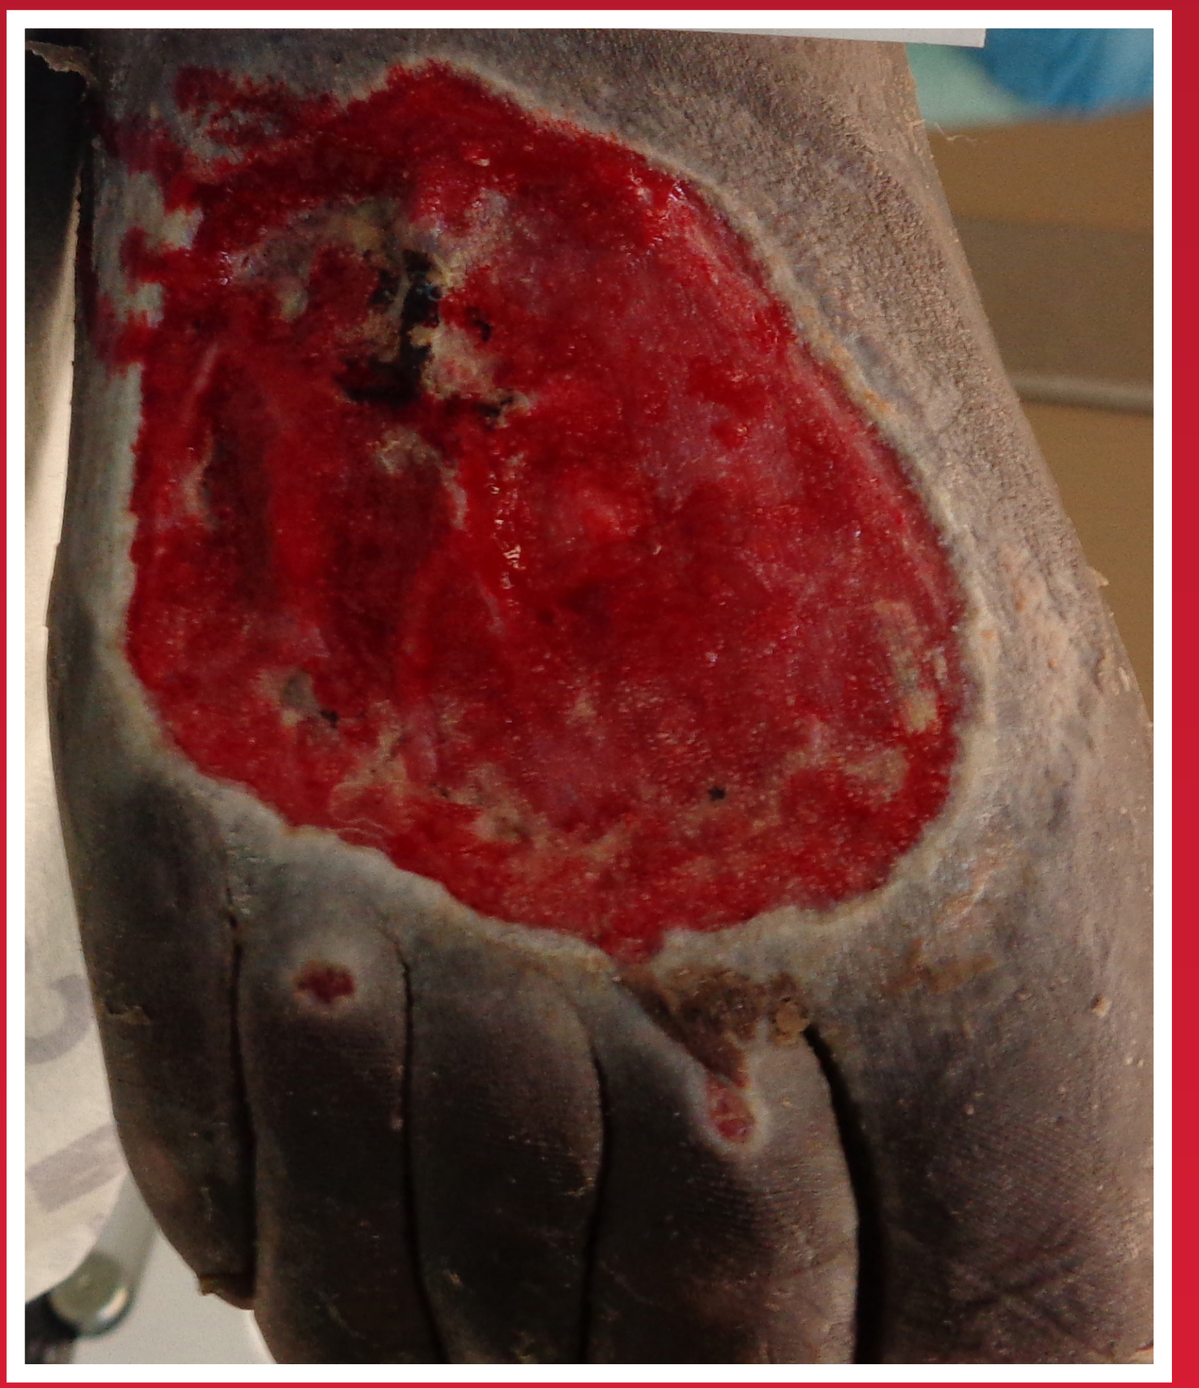

Foot wound week 2

RESULTS

- From the 25 patients enrolled, 18 reached the therapy goal within the study duration. Seven patients terminated the therapy early, with 5 achieving acceptable progress.
- Therapy goals reached for all 18 patients that completed the study in a mean duration of 20 ± 5 days. Outcomes comparable to previous data using the same single-patient-use NPWT system* [1, 2] and with other literature on NPWT treated patients with similar wound indications [3].
- Clinicians were satisfied with the ease of use, dressing change, the intuitive device operation and the dressing seal. Furthermore, patient satisfaction and mobility was perceived very well.

SUBJECT	WOUND TYPE	LOCATION
DOCTORS HOSPITAL OF AUGUSTA		
001	Pilonidal Cyst	Sacrum
002	Diabetic Foot Ulcer	Right Foot
003	Pressure Ulcer	Sacrum
004	Abcess	Right Groin
005	Full Thickness Wound	Left Flank
006	Carbuncle	Right Buttock
007	Dehisced Surgical Wound	Abdomen
008	Pilonidal Cyst	Sacrum
009	Abcess	Abdomen
010	Diabetic Foot Ulcer	Left Foot
011	Full Thickness Wound	Back
012	Necrotizing Fasciitis	Right Thigh
013	Post Amputation Wound	Right Stump
014	Diabetic Foot Ulcer	Right Foot
015	Full Thickness Wound	Left Thigh
016	Necotizing Soft Tissue Infection	Right Foot
017	Diabetic Foot Ulcer	Left Heel
018	Diabetic Foot Ulcer	Right Foot
WELLSTAR COBB HOSPITAL		
001	Hematoma	Left Lower Leg
002	Full Thickness Wound	Abdomen
003	Non-Healing Surgical Wound	Abdomen
004	Necrotizing Fasciitis	Upper Back
005	Abcess	Lower Back
006	Necrotizing Soft Tissue Infection	Abdomen
007	Full Thickness Wound	Abdomen

**For all wounds arterial etiology was ruled out*

CONCLUSION

This study confirms earlier evidence that the use of a small portable lightweight personal use NPWT system* supports wound healing for a wide variety of wounds of various sizes through its capacity to accurately deliver set pressure to the wound and efficiently remove exudate. It enables patients to be treated at home, which gains increasing importance in pandemic times, and is easy to handle and operate for clinicians.

NOTES

The Joseph M. Still Research Foundation, Inc., received financial support for meeting registrations for the presenter by Medela. Pat Schwab is employed by Medela LLC. None of the other authors have any other disclosures to make related to the sponsor.

Product notation: *Invia® Motion™ NPWT system/Invia® NPWT Dressings | **Invia® FitPad
Acknowledgements: The support of Medela AG (Laettichstrasse 4b, 6340 Baar, Switzerland) for this project is gratefully acknowledged.
Trademarks: Medela, Invia and Invia Motion are registered in the U.S. Patent and Trademark Office and elsewhere.

REFERENCES

[1] Inpatient Surgical Setting Transition to Outpatient: Supportive Role of a New Personal and a Reusable Negative Pressure Wound Therapy (NPWT) System with Double Lumen. Presented at the Annual Symposium on Advanced Wound Care (SAWC) Spring, April 25 - 29, 2018, Charlotte, NC, USA
[2] Evaluating the clinical acceptance of a Negative Pressure Wound Therapy (NPWT) system in accordance with standards of care in patients with a variety of wound types. Presented at the Annual Symposium on Advanced Wound Care (SAWC) Spring, May 13-17, 2020, San Diego, CA, USA
[3] Negative-pressure wound therapy for management of chronic neuropathic noninfected diabetic foot ulcerations - short-term efficacy and long-term outcomes. Borys et. al., Endocrine. 2018 Dec;62(3):611-616. doi: 10.1007/s12020-018-1707-0. Epub 2018 Aug 11

JOSEPH M. STILL RESEARCH FOUNDATION | BRCA BURN AND RECONSTRUCTIVE CENTERS OF AMERICA | DOCTORS HOSPITAL Joseph M. Still Burn Center | Wellstar